P2047

Time-Kill Kinetics and Post-Antibiotic Effect of Cadazolid against *Clostridium difficile*

Kirsty.Skinner@evotec.com

*K. Skinner[1], S. Birchall[1], D. Corbett[1], P. Thommes[1], H. H. Locher[2]
[1]Evotec (UK) Ltd, Manchester, United Kingdom, [2]Actelion Pharmaceuticals Ltd, Gewerbestrasse 16, 4123 Allschwil, Switzerland

INTRODUCTION

Over the past decade, *Clostridium difficile* has become a leading cause of nosocomial diarrhoea worldwide. This has been attributed, at least partly, to the emergence of hypervirulent *C. difficile* strains, such as ribotypes 027 and 078, which display increased resistance to a number of antibiotics and produce binary toxin in addition to toxin A and B (1). Infection caused by these strains has been associated with increased disease severity and mortality (2).

Cadazolid is a novel antibiotic which combines quinolone and oxazolidinone moieties into a new class of antibacterial agents referred to here as quinoxolidinones and is in clinical development for the treatment of *C. difficile* associated diarrhoea (CDAD), also known as *C. difficile* infection. In previous studies cadazolid showed potent *in vitro* activity against *C. difficile* clinical isolates (3-7), and in a human gut model of CDAD, while having only a very limited impact on bacteria of the normal gut microflora (7).

This current study was undertaken to determine the *in vitro* time-kill kinetic activity and post-antibiotic effect (PAE) of cadazolid in comparison to fidaxomicin and vancomycin against a panel of 4 *C. difficile* strains, representing isolates from ribotypes 027, 078, 087 and 001.

METHODS

- Antibacterial compounds: Cadazolid (ACT-179811) (Actelion Pharmaceuticals Ltd) and vancomycin (Alfa Aesar, Lot #W08A008) powder stocks were stored at 4°C, while fidaxomicin (Santa Cruz Biotechnology, Lot #J0213) was stored at -20°C as recommended by each manufacturer.
- Antibiotic susceptibility testing was performed by the broth microdilution method in pre-reduced BHIS medium (Oxoid brain heart infusion broth supplemented with 5g/L yeast extract and 0.025% L-cysteine), for consistency with later experiments. *C. difficile* ATCC 700057 was used as a reference control (8).
- Time-kill kinetics, to investigate rate of killing for each antibiotic at sub- and supra-MIC concentrations, and PAE experiments, to evaluate the delayed regrowth of strains following 1h exposure to each antibiotic, were performed as previously described, with some modifications (9).
- Time-kill and PAE experiments were performed in triplicate on separate days testing cadazolid, fidaxomicin and vancomycin at concentrations of 0.5, 1, 2, 4, 8 or 16x the MIC with total viable counts being determined at T-1h (PAE only) and at 0, 1, 2, 3 (time-kill only), 4, 6, 8, 24 and 48h post-exposure.
- The PAE was calculated using the equation PAE = *T* - *C*, where *T* represents the time (h) required for the viable count to increase ≥10-fold over the post-washing count in the presence of antibiotic, and *C* represents the time required for the viable count to increase ≥10-fold over the post-washing count in the absence of antibiotic (10).
- Bactericidal activity refers to ≥3 $\log_{10}$ reduction in viability relative to the starting inoculum after 24h exposure to test articles. The limit of detection (LOD) for these assays was 50 colony forming units (CFU)/mL.

RESULTS

Broth Microdilution Method

- The MICs of cadazolid and fidaxomicin for the *C. difficile* strains tested by the broth microdilution method were 0.125 to 0.25 µg/mL, and 0.008 to 0.25 µg/mL, respectively, both of which were within published ranges (4, 12).
- Vancomycin susceptibility for the 4 *C. difficile* isolates and *C. difficile* ATCC 700057 reference control were within the CLSI-suggested MIC range of 0.5 to 4 µg/mL (11).

Table 1. Cadazolid and comparator antibiotic modal MICs determined by broth microdilution against 4 *C. difficile* isolates

C. difficile strain	Ribotype	Antibiotic (MIC; µg/mL)		
		Cadazolid	Vancomycin	Fidaxomicin
ATCC 43255	087	**0.25**	2	0.25
NCTC 13366	027	**0.125**	1	0.25
ATCC BAA-1875	078	**0.25**	2	0.125
ATCC 9689	001	**0.25**	2	0.008

Time-kill kinetics

- Cadazolid achieved bactericidal activity (≥3 $\log_{10}$ CFU/mL reduction) against *C. difficile* ATCC 43255, NCTC 13366 (ribotype 027) (Fig. 1; Table 2) and ATCC BAA-1875 (ribotype 078) (Fig. 1), while with ATCC 9689 a 2-3 $\log_{10}$ reduction in CFU/mL was achieved (Table 2).
- Cadazolid killing rates were concentration-dependent in a range of 0.5 to 2-fold the MIC, but were generally not further increased at concentrations of ≥4-fold the MIC. Increasing the exposure time from 24h to 48h only modestly increased the extent of killing by cadazolid (Fig. 1; Table 2).
- Vancomycin showed reduced initial killing rates and was bacteriostatic only at 24h in 3 out of 4 strains (Fig. 2; Table 2).
- Fidaxomicin showed comparable bactericidal effects to that of cadazolid, with some strain-to-strain variation (Fig. 3; Table 2).

Figure 1. Killing kinetics of cadazolid against *C. difficile* ATCC BAA-1875 (ribotype 078) (A) and NCTC 13366 (ribotype 027) (B).

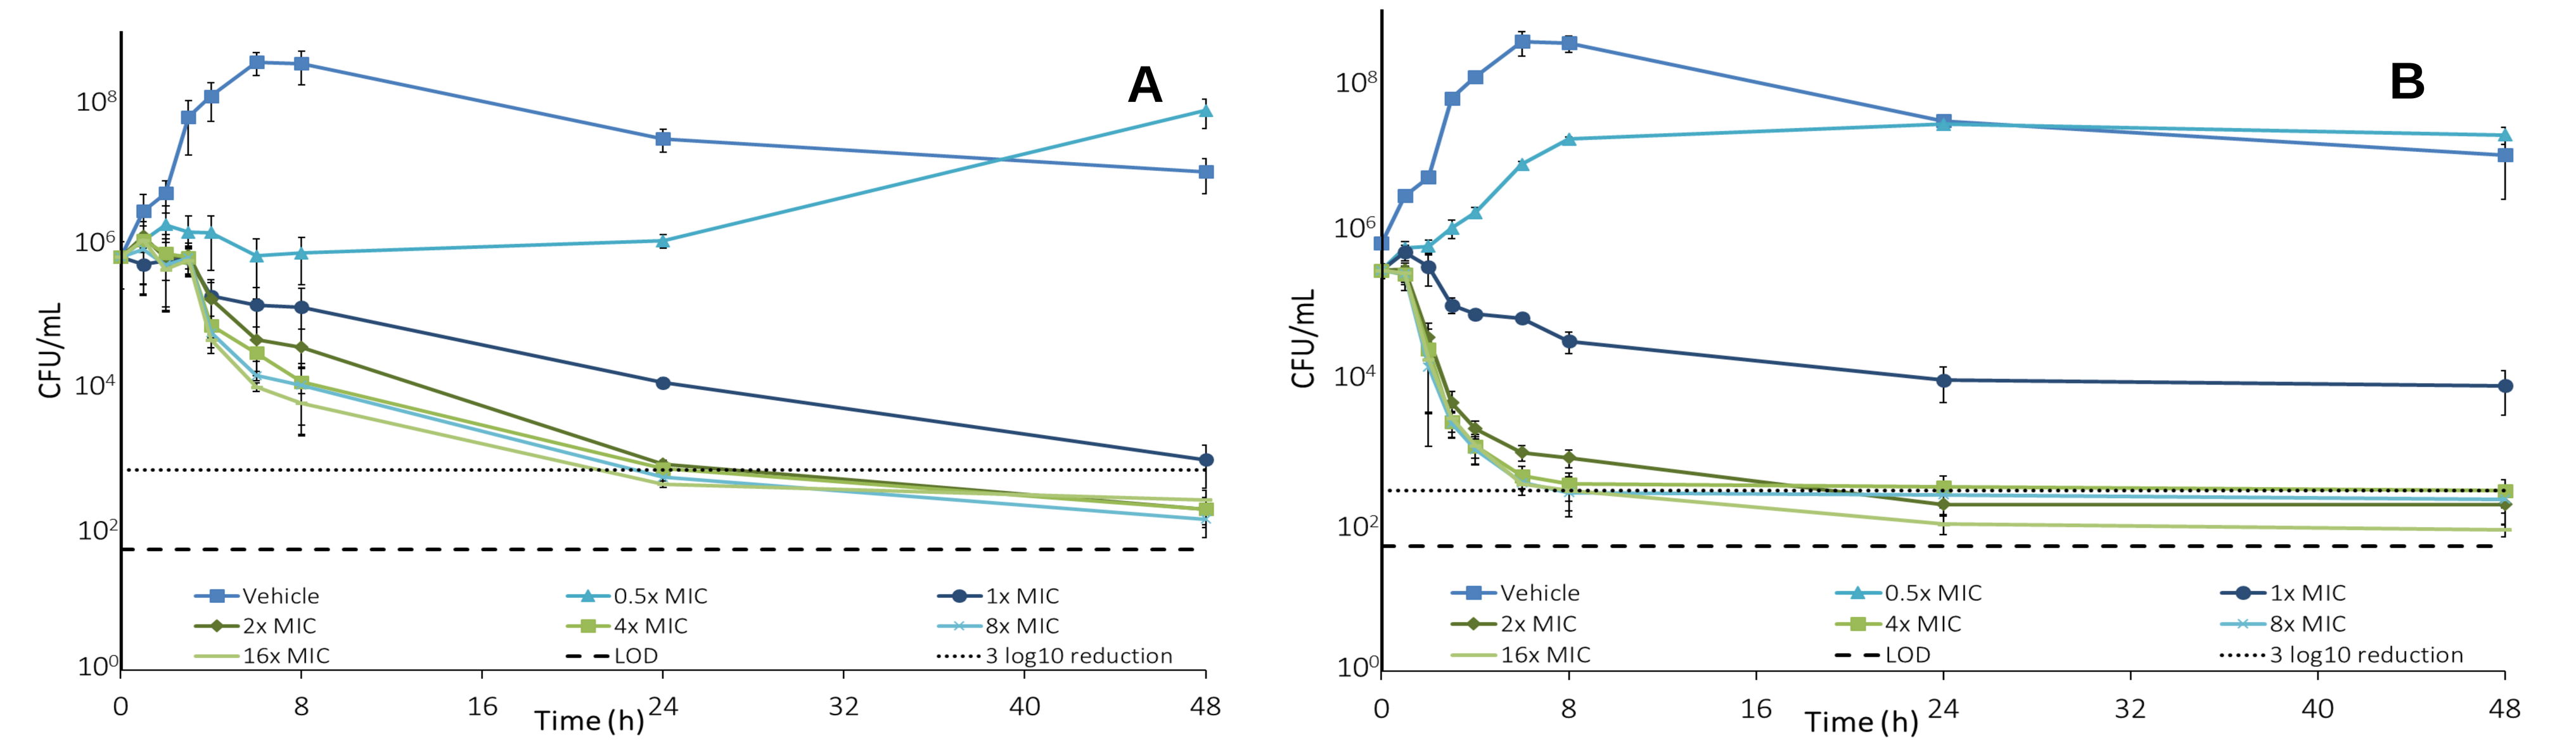
A
B
CFU/mL
Time (h)
Vehicle
0.5x MIC
1x MIC
2x MIC
4x MIC
8x MIC
16x MIC
LOD
3 log10 reduction

Figure 2. Killing kinetics of vancomycin against *C. difficile* ATCC BAA-1875 (ribotype 078) (A) and NCTC 13366 (ribotype 027) (B).

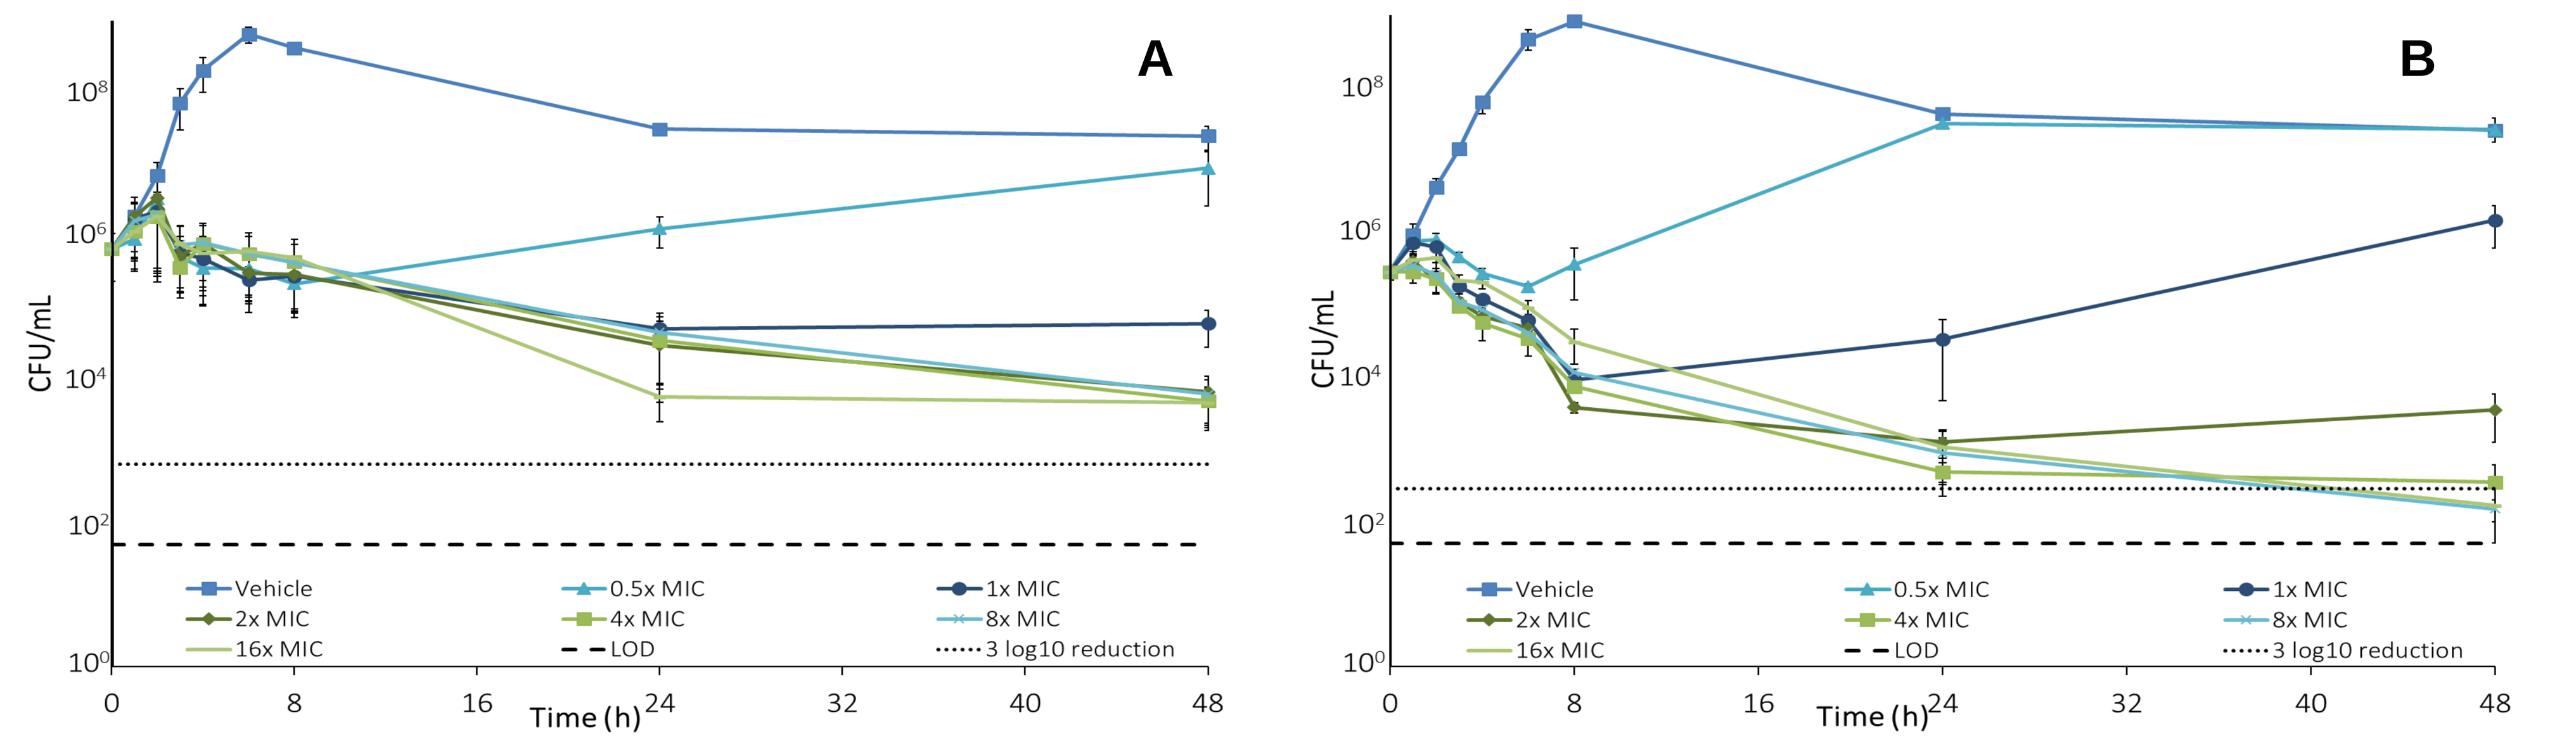
A
B
CFU/mL
Time (h)
Vehicle
0.5x MIC
1x MIC
2x MIC
4x MIC
8x MIC
16x MIC
LOD
3 log10 reduction

Figure 3. Killing kinetics of fidaxomicin against C. difficile ATCC BAA-1875 (ribotype 078) (A) and NCTC 13366 (ribotype 027) (B).

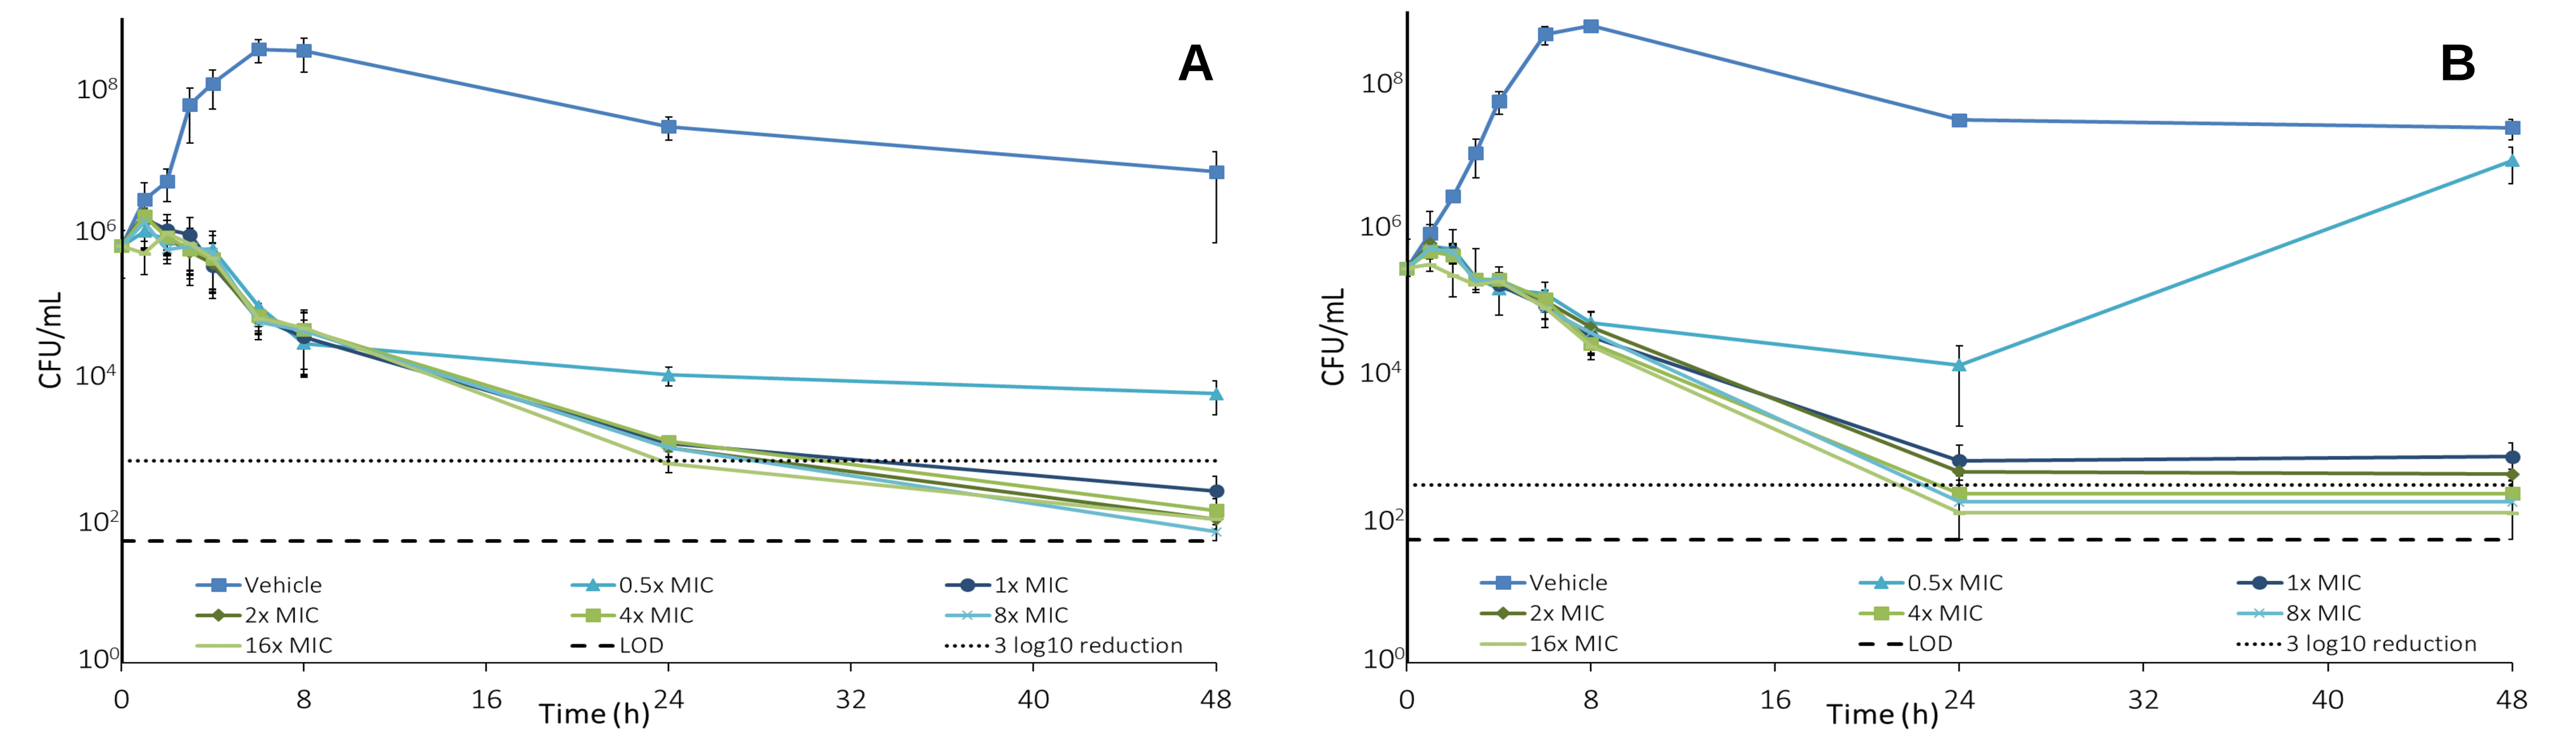
A
B
CFU/mL
Time (h)
Vehicle
0.5x MIC
1x MIC
2x MIC
4x MIC
8x MIC
16x MIC
LOD
3 log10 reduction

Table 2. $\log_{10}$ CFU/mL reduction values for key time points and antibiotic concentrations against 4 *C. difficile* isolates

C. difficile strain	Agent	CFU/mL $\log_{10}$ reduction at 24 h		CFU/mL $\log_{10}$ reduction at 48 h	
		4x MIC	16x MIC	4x MIC	16x MIC
ATCC 43255	**Cadazolid**	**3.82±0.76**	**4.18±0.84**	**4.13±0.21**	**3.75±0.53**
	Vancomycin	3.00±0.49	2.71±0.52	2.19±1.25	4.13±0.24
	Fidaxomicin	2.47±0.95	2.27±0.90	3.19±0.78	3.82±0.76
NCTC 13366	**Cadazolid**	**3.09±0.34**	**3.46±0.09**	**3.00±0.18**	**3.52±0.04**
	Vancomycin	2.87±0.43	2.60±0.41	2.91±0.82	3.42±0.45
	Fidaxomicin	3.38±0.53	3.49±0.35	3.12±0.53	3.49±0.35
ATCC BAA-1875	**Cadazolid**	**2.82±0.38**	**3.01±0.56**	**3.55±0.20**	**3.29±0.40**
	Vancomycin	1.69±0.79	2.00±0.85	2.12±1.15	2.19±0.51
	Fidaxomicin	2.56±0.35	2.88±0.35	3.69±0.31	3.67±0.40
ATCC 9689	**Cadazolid**	**2.53±0.19**	**1.74±0.07**	**2.91±0.23**	**2.31±0.51**
	Vancomycin	2.70±0.04	1.67±0.18	3.77±0.26	3.37±0.18
	Fidaxomicin	2.60±0.26	3.23±0.73	2.96±0.19	3.65±0.27

Post antibiotic effect (PAE)

- PAEs of cadazolid were short (0-2h) at concentrations up to 4-fold the MIC in all strains, however, prolonged PAEs (4->20h) were measured at 8 or 16x the MIC for *C. difficile* ATCC 43255 and ATCC BAA-1875 (Table 3).
- Vancomycin showed short PAEs (0-2h) with all strains and at all test concentrations (Table 3).
- Fidaxomicin showed prolonged PAEs (6->20h) at 4-fold the MIC in 3 out of 4 strains and in all strains at 16-fold the MIC in agreement with published data (Table 3; 10, 12).

Table 3. PAEs following 1h exposure to cadazolid, vancomycin or fidaxomicin against 4 *C. difficile* isolates

C. difficile strain	Agent	PAE (h)					
		0.5x MIC	1x MIC	2x MIC	4x MIC	8x MIC	16x MIC
ATCC 43255	**Cadazolid**	**0**	**0**	**0**	**0**	**20**	**>20**
	Vancomycin	0-2	0-2	0-2	0	0-2	0-2
	Fidaxomicin	0-2	4	8-20	8-20	8-20	>20
NCTC 13366	**Cadazolid**	**2**	**2**	**0**	**0**	**0**	**2**
	Vancomycin	2	2	2	2	2	2
	Fidaxomicin	2	4	4	6	8-22	8-22
ATCC BAA-1875	**Cadazolid**	**2**	**2**	**2**	**2**	**4**	**8-22**
	Vancomycin	0	0	2	2	2	2
	Fidaxomicin	4	6	8-22	8-22	8-22	8-22
ATCC 9689	**Cadazolid**	**2**	**2**	**2**	**2**	**2**	**2**
	Vancomycin	2	2	2	2	2	2
	Fidaxomicin	2	2	2	2	2	8-22

CONCLUSIONS

- **Cadazolid displayed potent *in vitro* activity against all of the *C. difficile* isolates tested, generating low MICs consistent with recently published literature (3-7).**
- **Cadazolid-mediated killing was faster and occurred at lower concentrations than observed for vancomycin, while potency and killing was comparable to that observed for fidaxomicin.**
- **Notably, cadazolid also displayed a potent bactericidal effect against fluoroquinolone-resistant hypervirulent ribotype 027 and 078 strains.**
- **PAEs of cadazolid varied depending on strain and test concentration.**

References

1. Martin JSH, et al. Nature Reviews Gastroenterology & Hepatology 2016;13:2016-16.
2. Rupnik M, et al. Nature Reviews Microbiology 2009;7:526-36.
3. Locher HH, et al. Antimicrob Agents Chemother 2014; 58:901-8.
4. Locher HH, et al. Antimicrob Agents Chemother 2014; 58: 892-900.
5. Rashid MU, et al. Anaerobe 2013; 20: 32-35.
6. Hecht DW, et al. Abstract/poster E-808 of the 52nd Interscience Conference on Antimicrobial Agents and Chemotherapy, 2012, San Francisco, American Society for Microbiology, Washington, DC, USA.
7. Chilton CH, et al. J Antimicrob Chemother 2014; 69:697-705.
8. CLSI document M11-A8, 2012, Clinical Laboratory Standards Institute, 940 West Valley Road, Suite 1400, Wayne, Pennsylvania, USA.
9. Corbett D, et al. Antimicrob Agents Chemother 2015; 70: 1751-1756.
10. Babakhani F, et al. Antimicrob Agents Chemother 2011; 55: 4427-4429.
11. CLSI document M100S. 2016, Clinical Laboratory Standards Institute, 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087, USA.
12. Goldstein EJC, et al. Clin Infect Dis, 2012; 55: S143-148.

Disclosures

This study was funded by Actelion Pharmaceuticals Ltd.

Presented at the 27th European Congress of Clinical Microbiology and Infectious Diseases (ECCMID), April 22-25 2017, Vienna, Austria.